Route to a safe delivery strategy of AAV-based gene therapy to the caudate and putamen of nonhuman primates

Francesca Cargnin, PhD[1]; Jeremy Brower, PhD[1]; Mark Johnson, PhD[2]; Christen Simon, PhD[2]; Andy Carlson, BS[2]; Haiyan Ma, PhD[3]; Smrithi Padmakumar, PhD[1]; Heena Beck, MS[1]; Edward Chroscinski, VMD[1]; Renee Gentzel, PhD[1]; Mohamad Nayal, MS[1]; Joan Wicks, DVM, PhD[1]; Brad Bolon, DVM, MS, PhD[4]; George Atkins, MS[1]; Xue Wang, PhD[5]; Charlie Li, PhD[1]; Mathew Li, PhD[1]; Barbara Clark, BS[1]; Alexandra Ellis, BS[1]; Drew Peterson, PhD[1]; Graciela Rivera-Pena, BS[1]; Phillip Price, PhD[1]; Christopher Cali, PhD[1]; JoAnn Coleman, MS[1]; Nino Devidze, PhD[1]; Elizabeth Ramsburg, PhD[1].

[1] Spark Therapeutics, Philadelphia PA; [2] Northern Biomedical Research, Norton Shores MI; [3] Northern Biomolecular Services, Portage MI; [4] GEMpath, Longmont CO; [5] Invicro, Boston MA

ABSTRACT

Adeno-associated virus (AAV) is effective for delivering genetic medicines to the brain through various routes, such as direct brain infusion, cerebrospinal fluid (CSF) compartment infusion, or intravenous infusion.

SPK-10001, a vectorized artificial microRNA aimed at treating Huntington's disease (HD), induces a dose-dependent reduction of both Huntingtin (HTT) mRNA and protein levels.

Since SPK-10001's proprietary capsid does not cross the blood-brain barrier via intravenous delivery, studies were conducted to find the safest and most effective administration route. Results showed that only direct injection into the brain parenchyma significantly reduced HTT protein in the caudate and putamen. Intra-CSF injection did not improve target coverage and could not replace intraparenchymal injection.

Initial injection methods caused mechanical and AAV-dose directed damage, but the optimization of the surgical technique improved both efficacy and safety outcomes. These optimizations allowed the administration of higher doses of SPK-10001 with minimal inflammatory response, highlighting the importance of surgical refinement in developing effective brain-directed gene therapies for human patients.

REFERENCES

1. Bates G.P. et al., Huntington disease. Nat.Rev.Dis. Primers. 2015; 1:15005
2. Sudhakar V. et al., J Neurosurg 133:530-537, 2020

ACKNOWLEDGMENTS.
The Authors are grateful for the unconditional support and collaboration demonstrated by their colleagues at Spark Therapeutics.
Illustrations created with BioRender.com

CONTACT

Francesca Cargnin, PhD
francesca.cargnin@sparktx.com

Liz Ramsburg, PhD
elizabeth.ramsburg@sparktx.com

Spark Therapeutics, Inc.

3025 / 3737 Market Street
Philadelphia, PA 19104

BACKGROUND

- HD is a rare neurodegenerative disorder, caused by an autosomal dominant repeat expansion (CAG) in the *HTT* gene, which leads to the production of mutant HTT protein (mHTT) on translation
- The toxic gain-of-function mechanism of mHTT drives nearly all cellular pathological mechanisms that underlie disease development (Bates et al., 2015[1])
- The striatum, and later the cerebral cortex, appears to be the brain region most susceptible to mHTT.
- Clinically, HD is characterized by progressive motor and cognitive impairment and behavioral dysfunction
- Reducing mHTT levels in the most affected brain regions could ameliorate all mHTT downstream pathogenic effects.

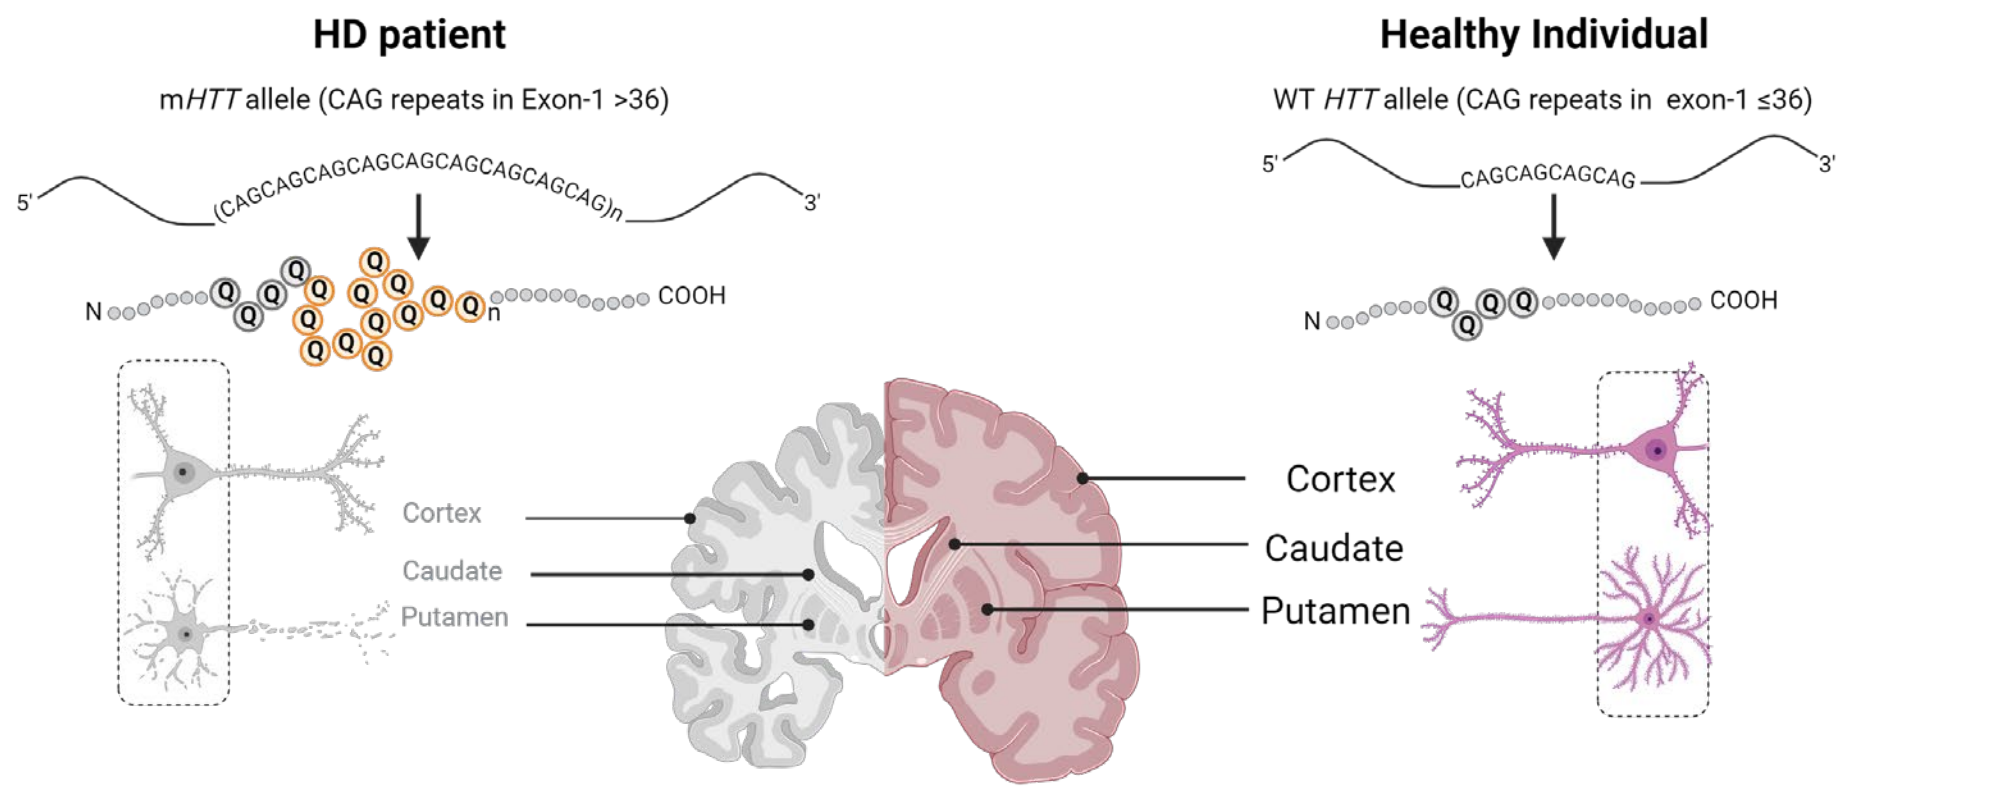
HD patient
m*HTT* allele (CAG repeats in Exon-1 >36)
Healthy Individual
WT *HTT* allele (CAG repeats in exon-1 ≤36)
COOH
Cortex
Caudate
Putamen

Figure 1. SCHEMATIC OF HUNTINGTON'S DISEASE ETIOLOGY AS RATIONALE TO DEVELOP A SURGICAL STRATEGY TO DELIVER GENE THERAPY TO THE BRAIN REGIONS OF CAUDATE AND PUTAMEN NUCLEI
mHTT is produced by every brain region but the most susceptible are the striatum (caudate and putamen) and later the cerebral cortex making the direct lowering of mHTT a potential disease-modifying therapy for Huntington's disease.

This study aimed to identify and optimize the surgical delivery to both the caudate and putamen, enhancing the safety and efficacy of AAV-mediated gene therapy treatments designed to reduce mHTT

EXPERIMENTAL APPROACH

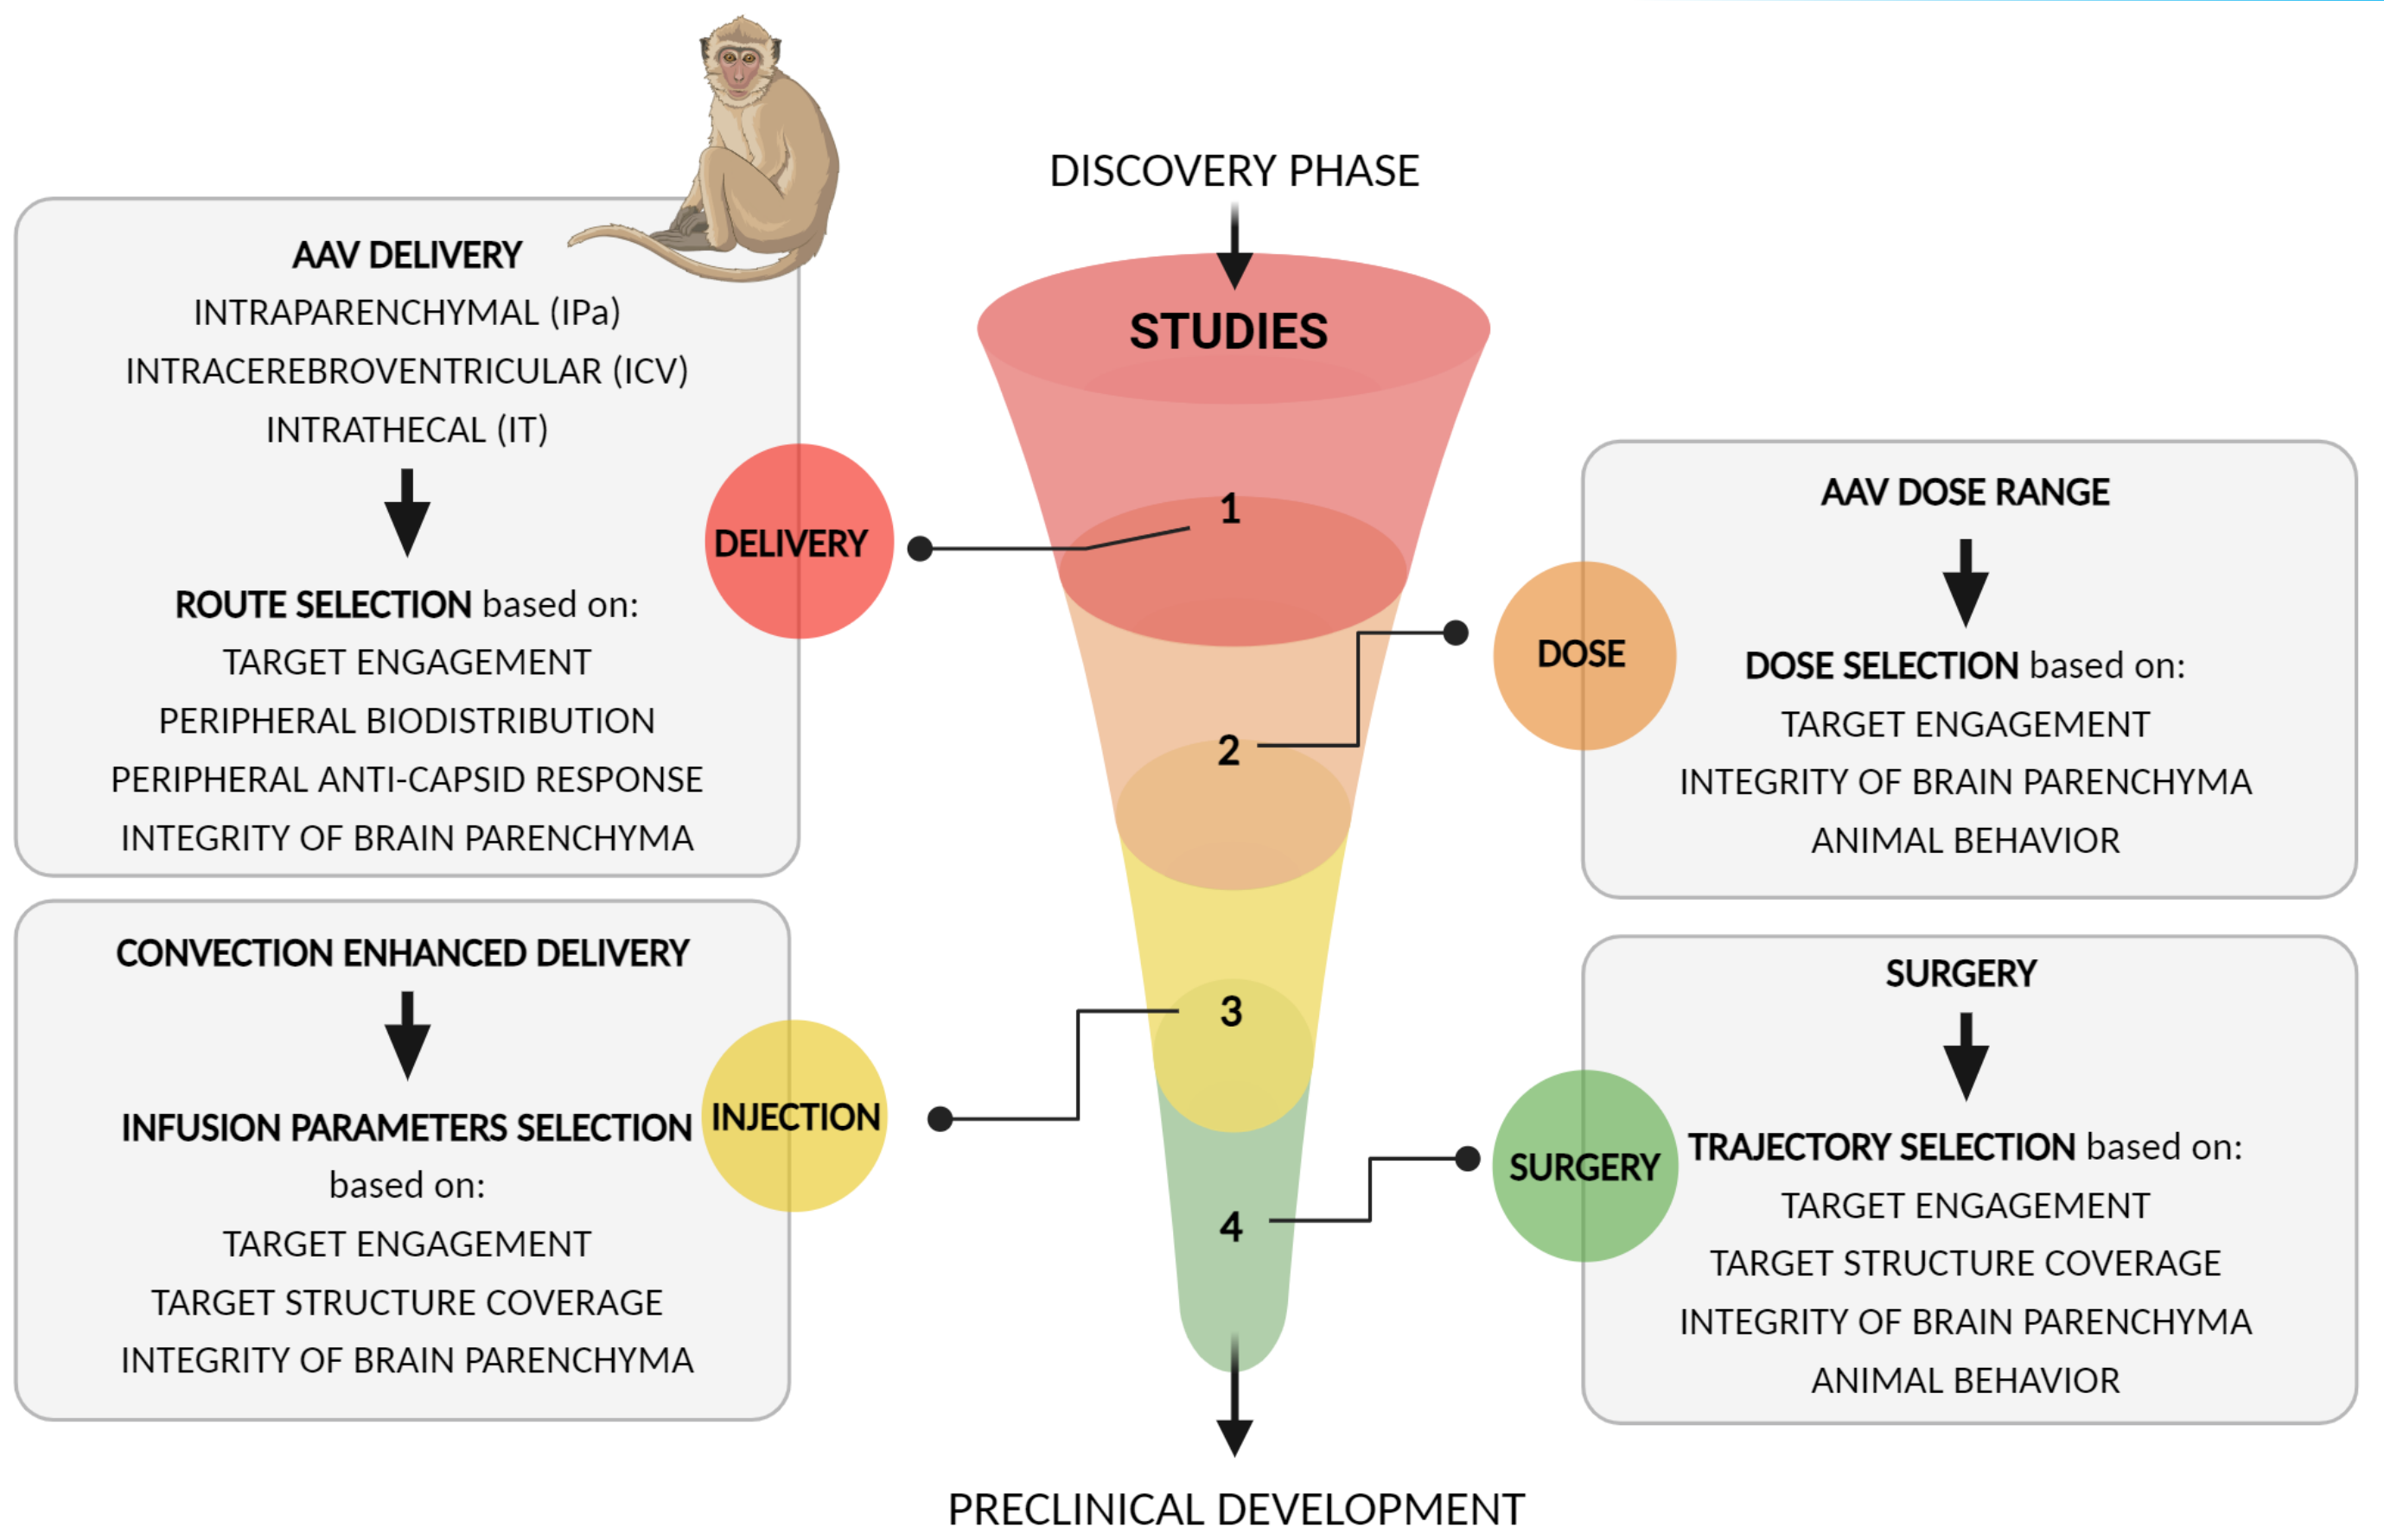
DISCOVERY PHASE
STUDIES
AAV DELIVERY
INTRAPARENCHYMAL (IPa)
INTRACEREBROVENTRICULAR (ICV)
INTRATHECAL (IT)
ROUTE SELECTION based on:
TARGET ENGAGEMENT
PERIPHERAL BIODISTRIBUTION
PERIPHERAL ANTI-CAPSID RESPONSE
INTEGRITY OF BRAIN PARENCHYMA
DELIVERY
1
AAV DOSE RANGE
DOSE
DOSE SELECTION based on:
TARGET ENGAGEMENT
INTEGRITY OF BRAIN PARENCHYMA
ANIMAL BEHAVIOR
2
CONVECTION ENHANCED DELIVERY
INFUSION PARAMETERS SELECTION based on:
TARGET ENGAGEMENT
TARGET STRUCTURE COVERAGE
INTEGRITY OF BRAIN PARENCHYMA
INJECTION
3
SURGERY
TRAJECTORY SELECTION based on:
TARGET ENGAGEMENT
TARGET STRUCTURE COVERAGE
INTEGRITY OF BRAIN PARENCHYMA
ANIMAL BEHAVIOR
4
PRECLINICAL DEVELOPMENT

Figure 2. SCHEMATIC OF EXPERIMENTAL PIPELINE TO ACHIEVE SAFE DELIVERY AND EFFICACY OF GENE THERAPY PRODUCTS.

RESULTS

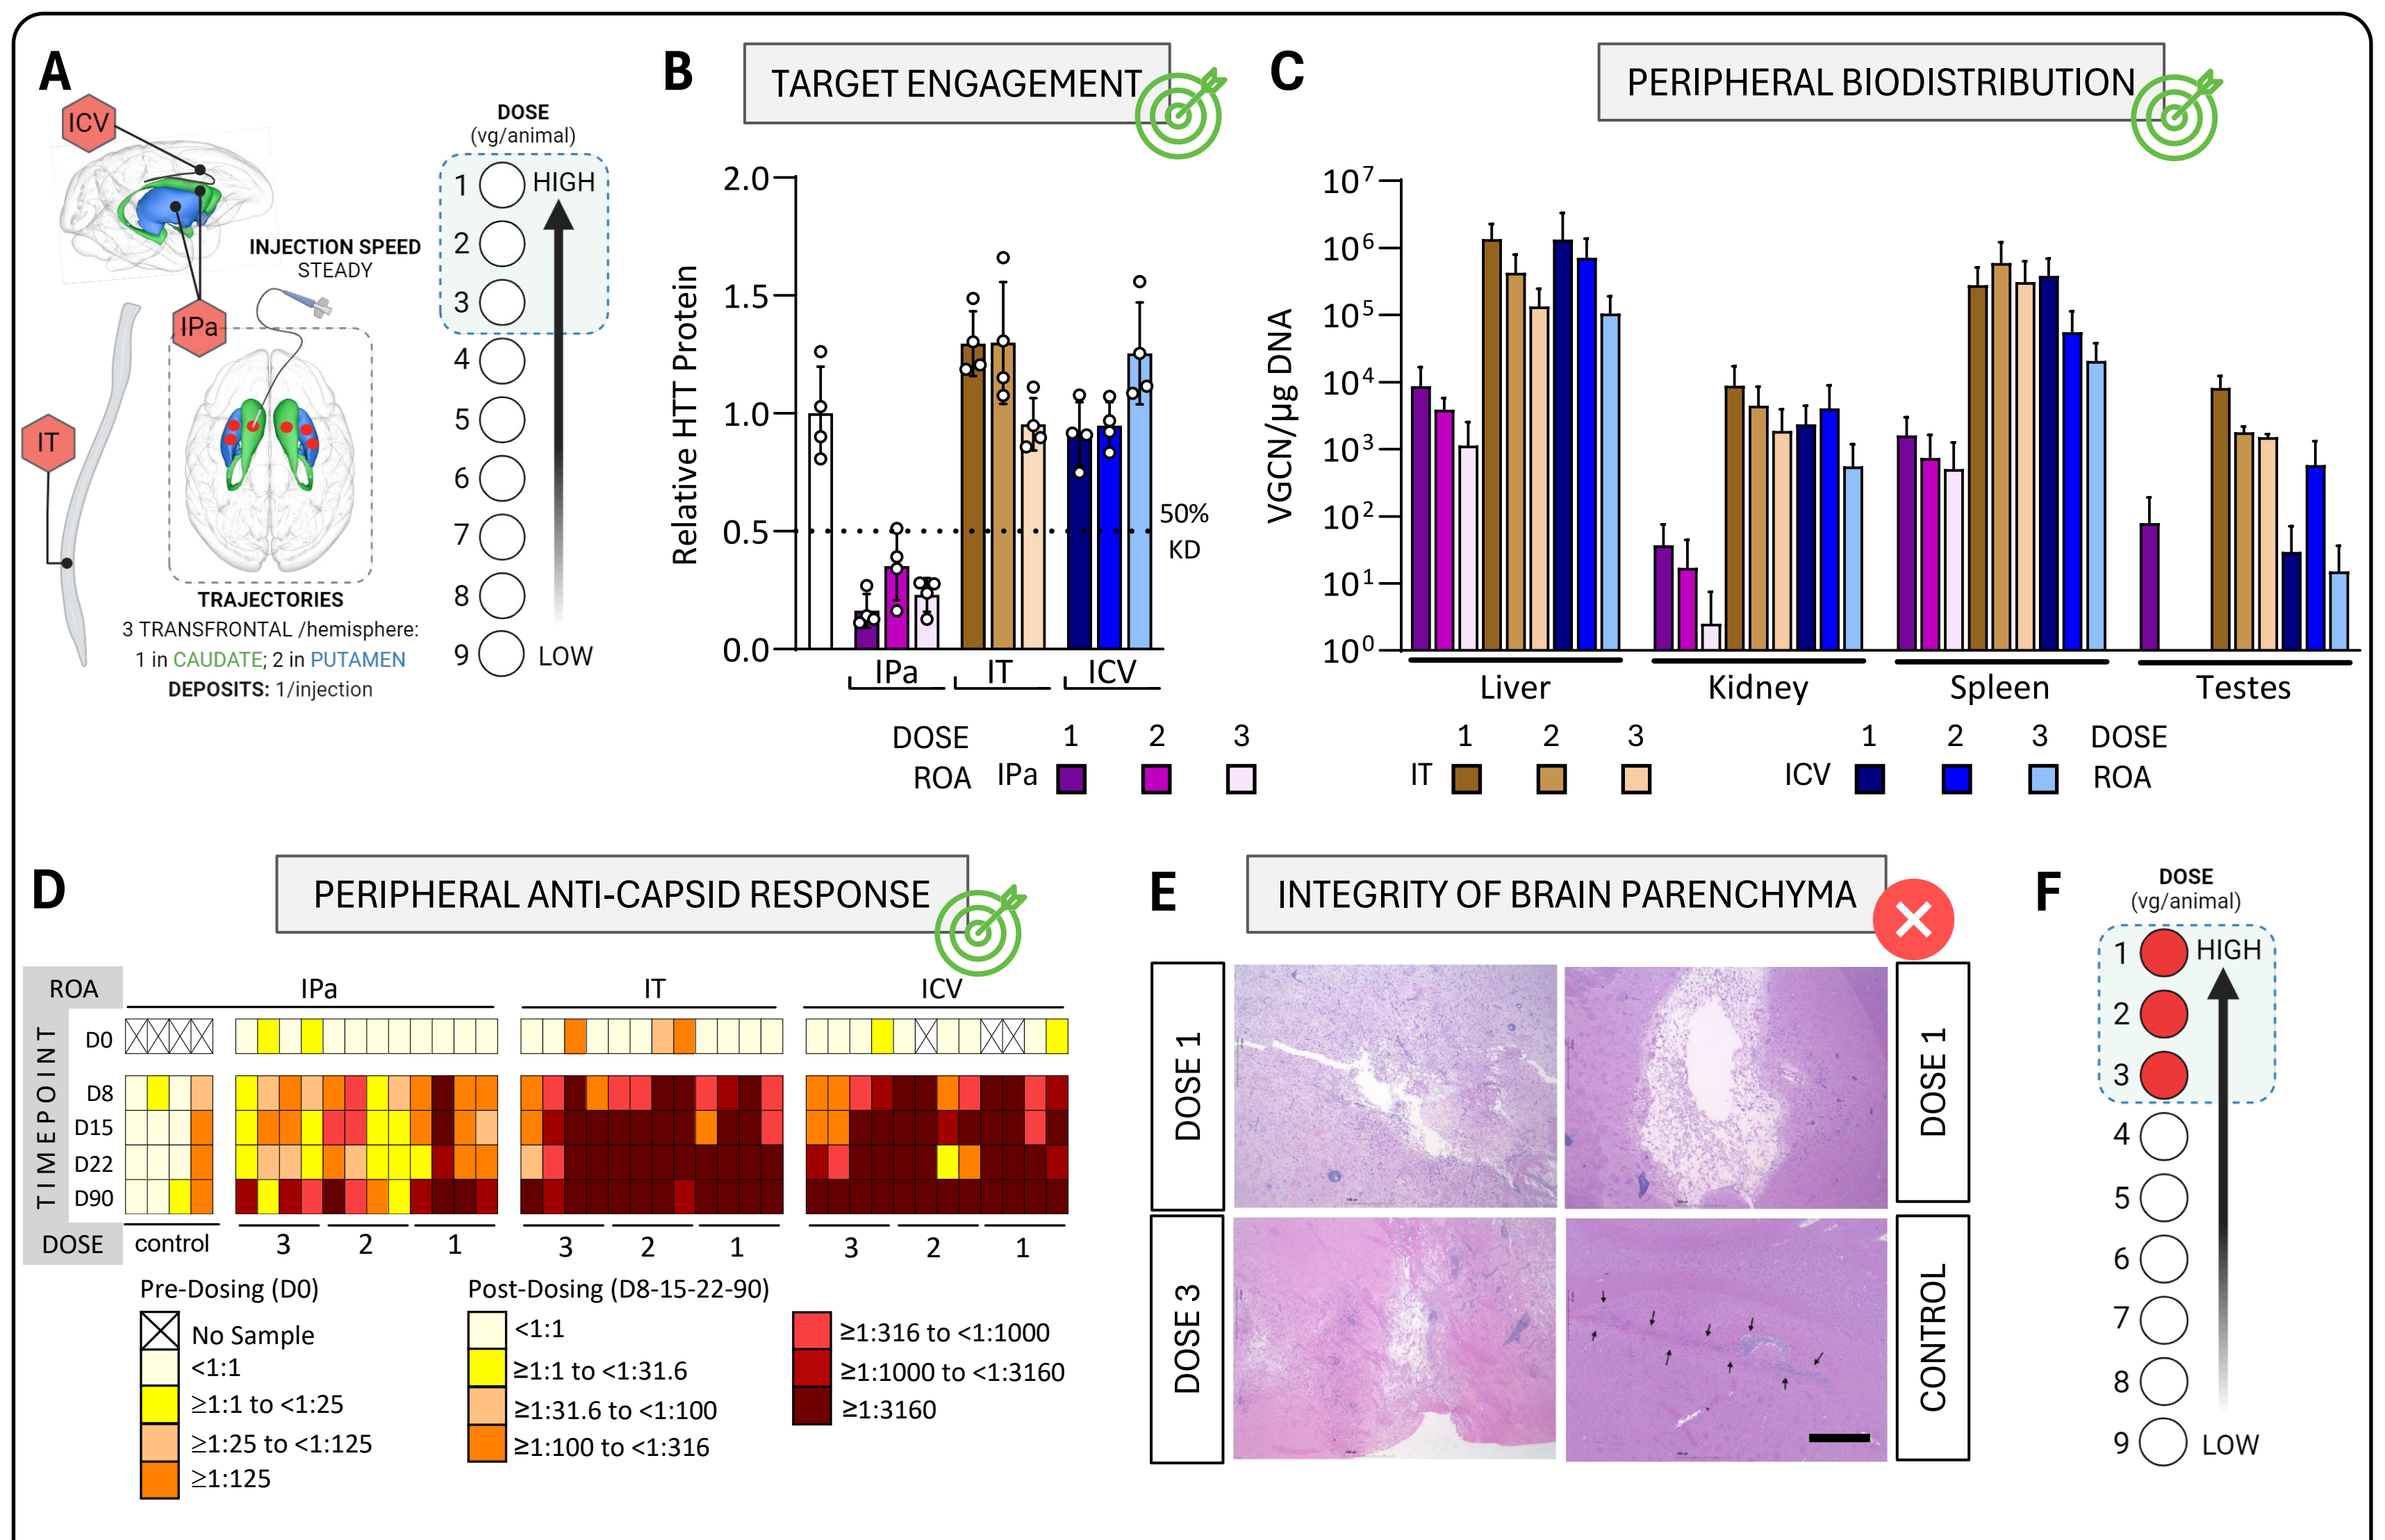
A
ICV
IPa
IT
INJECTION SPEED
STEADY
DOSE (vg/animal)
HIGH
LOW
TRAJECTORIES
3 TRANSFRONTAL /hemisphere:
1 in CAUDATE; 2 in PUTAMEN
DEPOSITS: 1/injection
B
TARGET ENGAGEMENT
Relative HTT Protein
50% KD
IPa
IT
ICV
DOSE
ROA
C
PERIPHERAL BIODISTRIBUTION
VGCN/μg DNA
Liver
Kidney
Spleen
Testes
D
PERIPHERAL ANTI-CAPSID RESPONSE
TIMEPOINT
D0
D8
D15
D22
D90
control
Pre-Dosing (D0)
No Sample
<1:1
≥1:1 to <1:25
≥1:25 to <1:125
≥1:125
Post-Dosing (D8-15-22-90)
<1:1
≥1:1 to <1:31.6
≥1:31.6 to <1:100
≥1:100 to <1:316
≥1:316 to <1:1000
≥1:1000 to <1:3160
≥1:3160
E
INTEGRITY OF BRAIN PARENCHYMA
DOSE 1
DOSE 3
CONTROL
F

STUDY 1. ROUTE OF ADMINISTRATION (ROA) SELECTION. IPa delivery of AAV effectively reduced HTT protein in NHP striatum with minimal spread to the periphery and moderate increase of anti-AAV NAb titer compared to other routes. However, histopathology analysis identified substantial pathological changes at all tested doses, suggesting further refinement of the dose range to decrease potential AAV-driven toxicity after IPa injections.
A. Schematic of the ROAs (IPa, IT, and ICV) and of the initial 3 AAV doses (1, 2, and 3, *blue box*) tested in Study 1. n=4 animals/group. **B.** HTT protein in the putamen of rhesus macaques 3 months after injection. Dotted line indicates 50% protein knockdown (KD) relative to control (*white bar*). **C.** Quantification of vector genome copy numbers (VGCN) in the periphery (representative organs). **D.** Anti-capsid Nab titers following AAV administration. **E.** Representative images of the histopathology assessment of the putamen after IPa injection. Mononuclear infiltrates, necrosis, and cavitation are visible in AAV-dosed tissues. *Arrows* indicate glial scar induced by mechanical damage in control animal. Scale bar=1 mm. **F.** Study 1 conclusions. *Red*= Effective dose but with adverse findings.

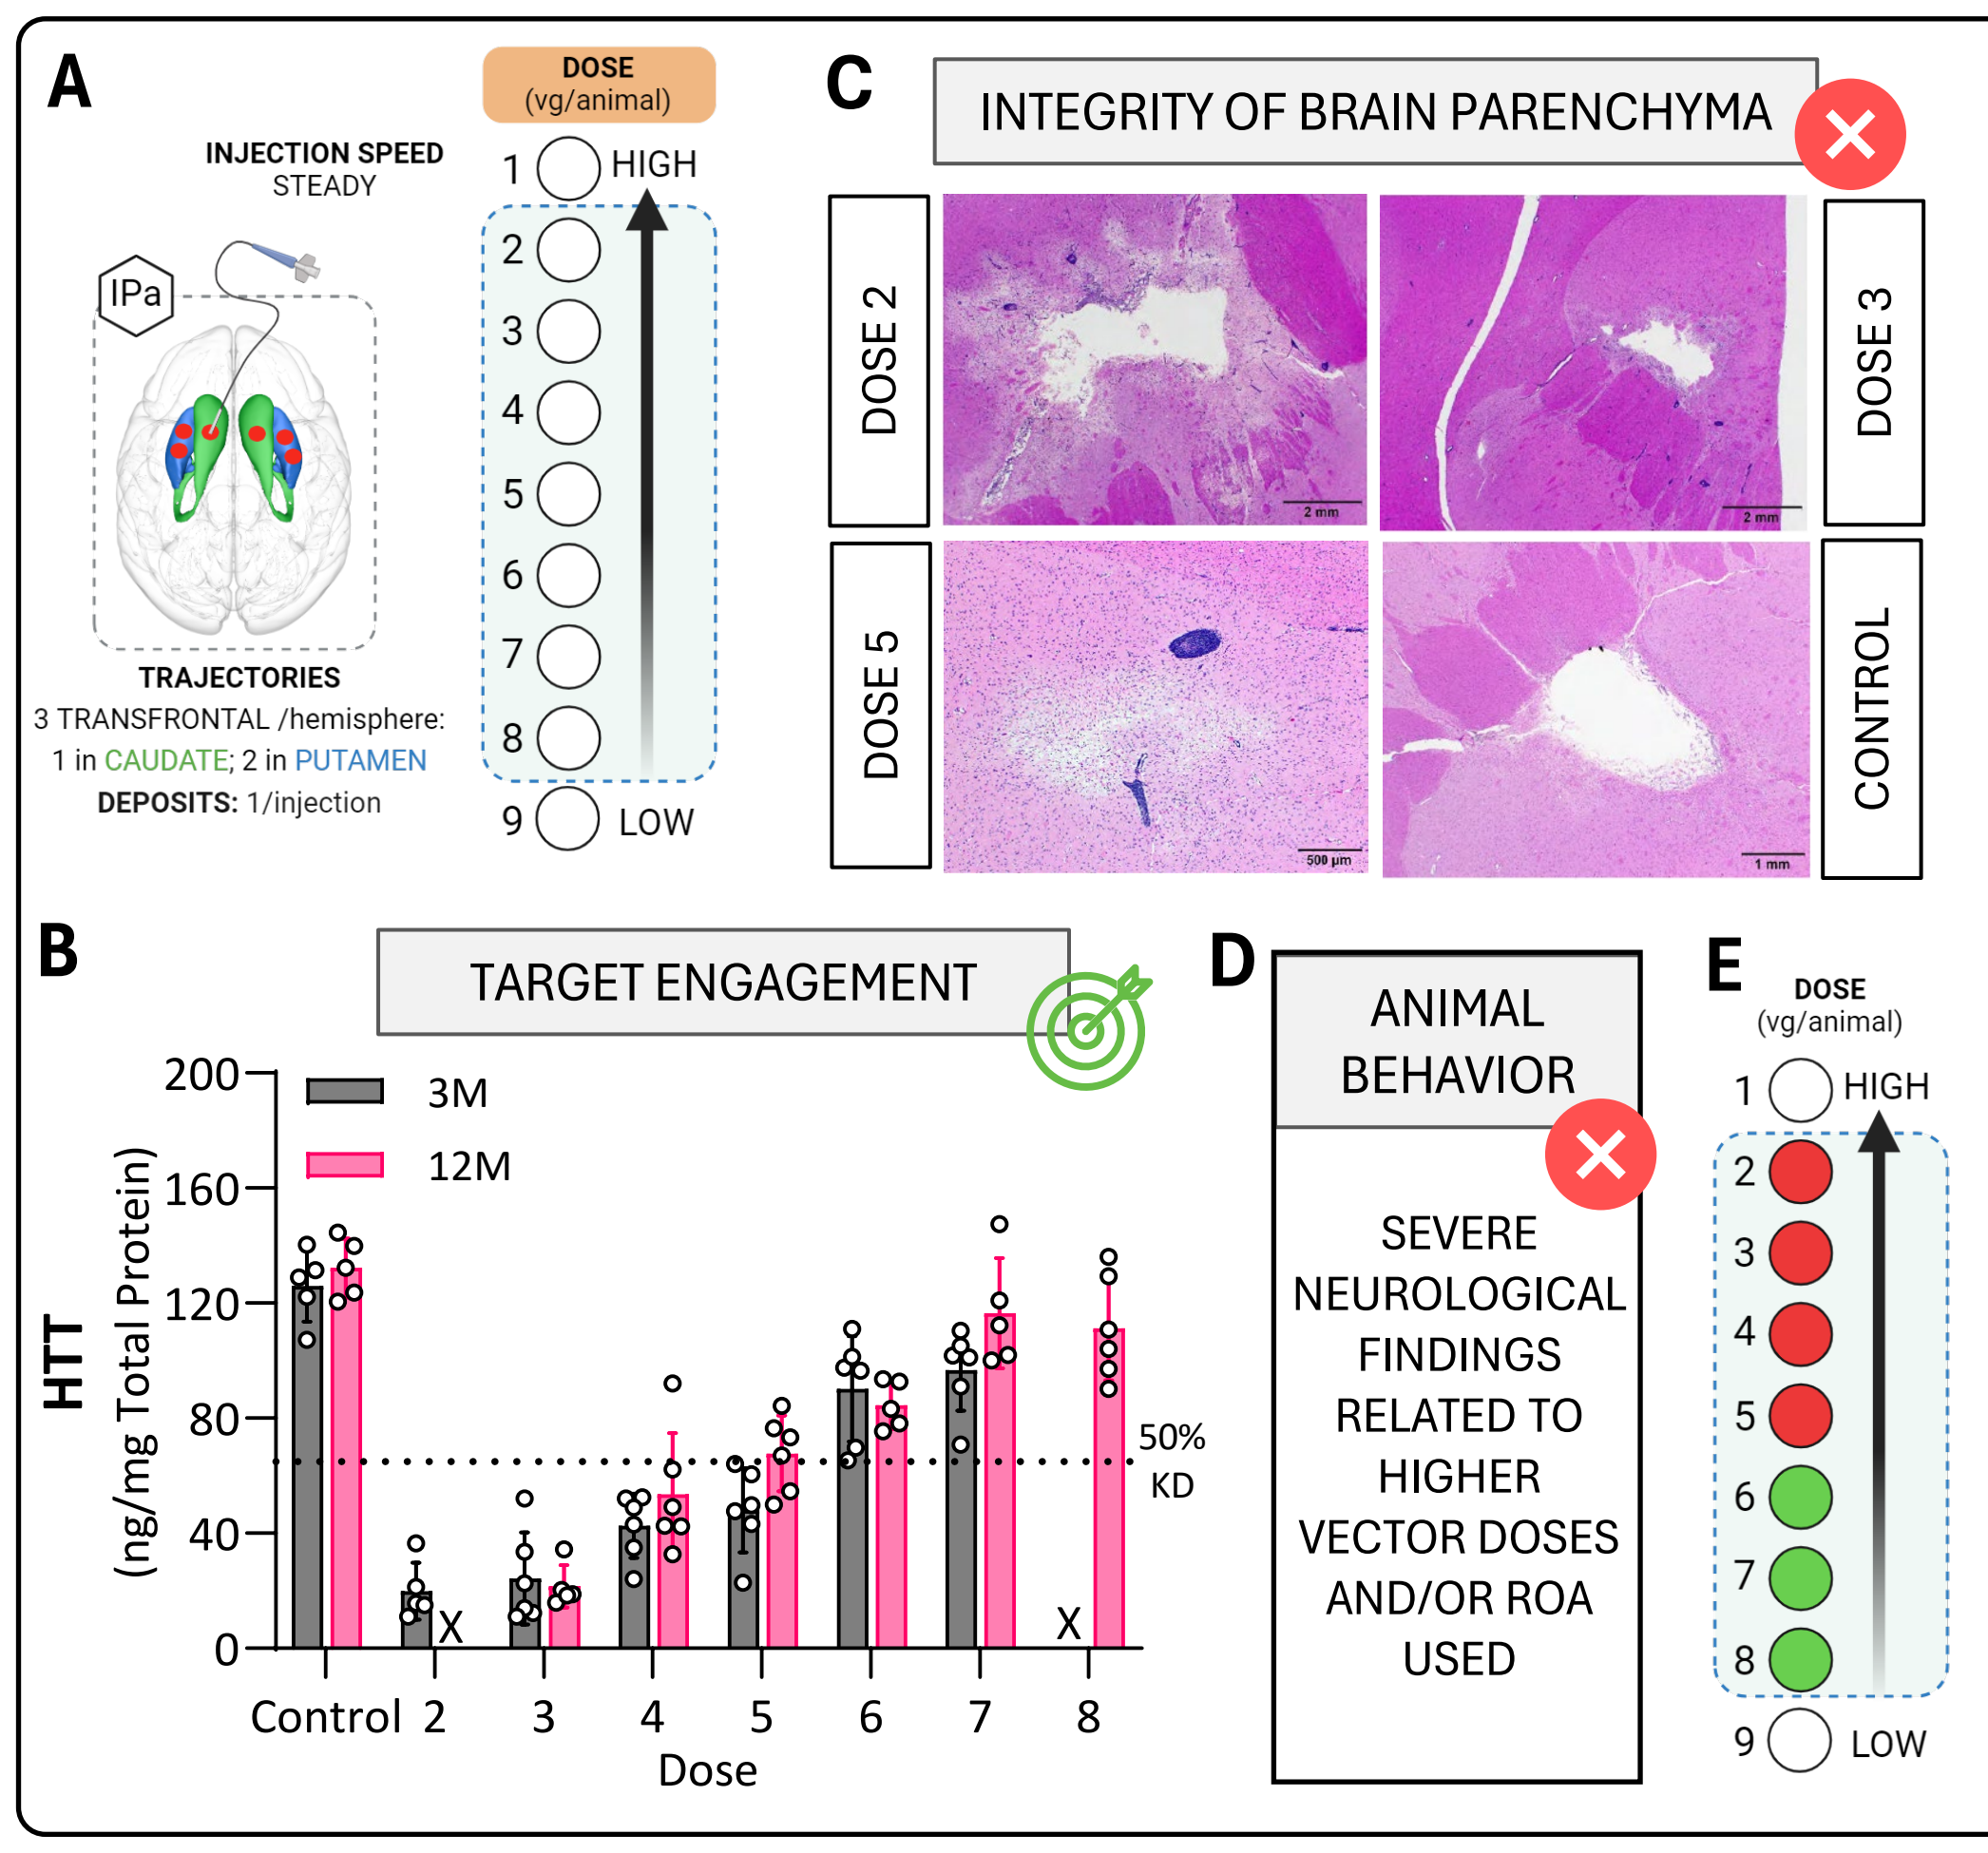
A
INJECTION SPEED
STEADY
DOSE (vg/animal)
HIGH
LOW
TRAJECTORIES
3 TRANSFRONTAL /hemisphere:
1 in CAUDATE; 2 in PUTAMEN
DEPOSITS: 1/injection
C
INTEGRITY OF BRAIN PARENCHYMA
DOSE 2
DOSE 3
DOSE 5
CONTROL
B
TARGET ENGAGEMENT
3M
12M
HTT (ng/mg Total Protein)
50% KD
Control
Dose
D
ANIMAL BEHAVIOR
SEVERE NEUROLOGICAL FINDINGS RELATED TO HIGHER VECTOR DOSES AND/OR ROA USED
E

STUDY 2. DOSE RANGE SELECTION. Expanding the dose range demonstrated that HTT protein could still be effectively reduced even after administering doses (4 to 7) lower than those tested in Study 1. However, histopathology analysis still identified substantial pathological changes associated with AAV-driven toxicity up to dose 5 and highlighted the impact of mechanical damage induced by IPa injection alone (control). Furthermore, the synergistic effect between brain parenchyma damage and high doses of AAV culminated in behavioral adverse events. This study suggested that both injection and surgical procedure had to be improved.
A. Schematic of the IPa injection procedure and of the 7 doses (from 2 to 8, *blue box*) tested in Study 2. n=6 animals/group. **B.** HTT protein in the putamen of rhesus macaques 3-12 months after injection. Dotted line indicates 50% protein knockdown (KD) relative to control. **C.** Representative images of the histopathology assessment of the putamen after IPa injection. **D.** Summary of behavioral adverse events. **E.** Study 2 conclusions. *Red*= Effective dose but with adverse findings. *Green*= effective and safe dose.

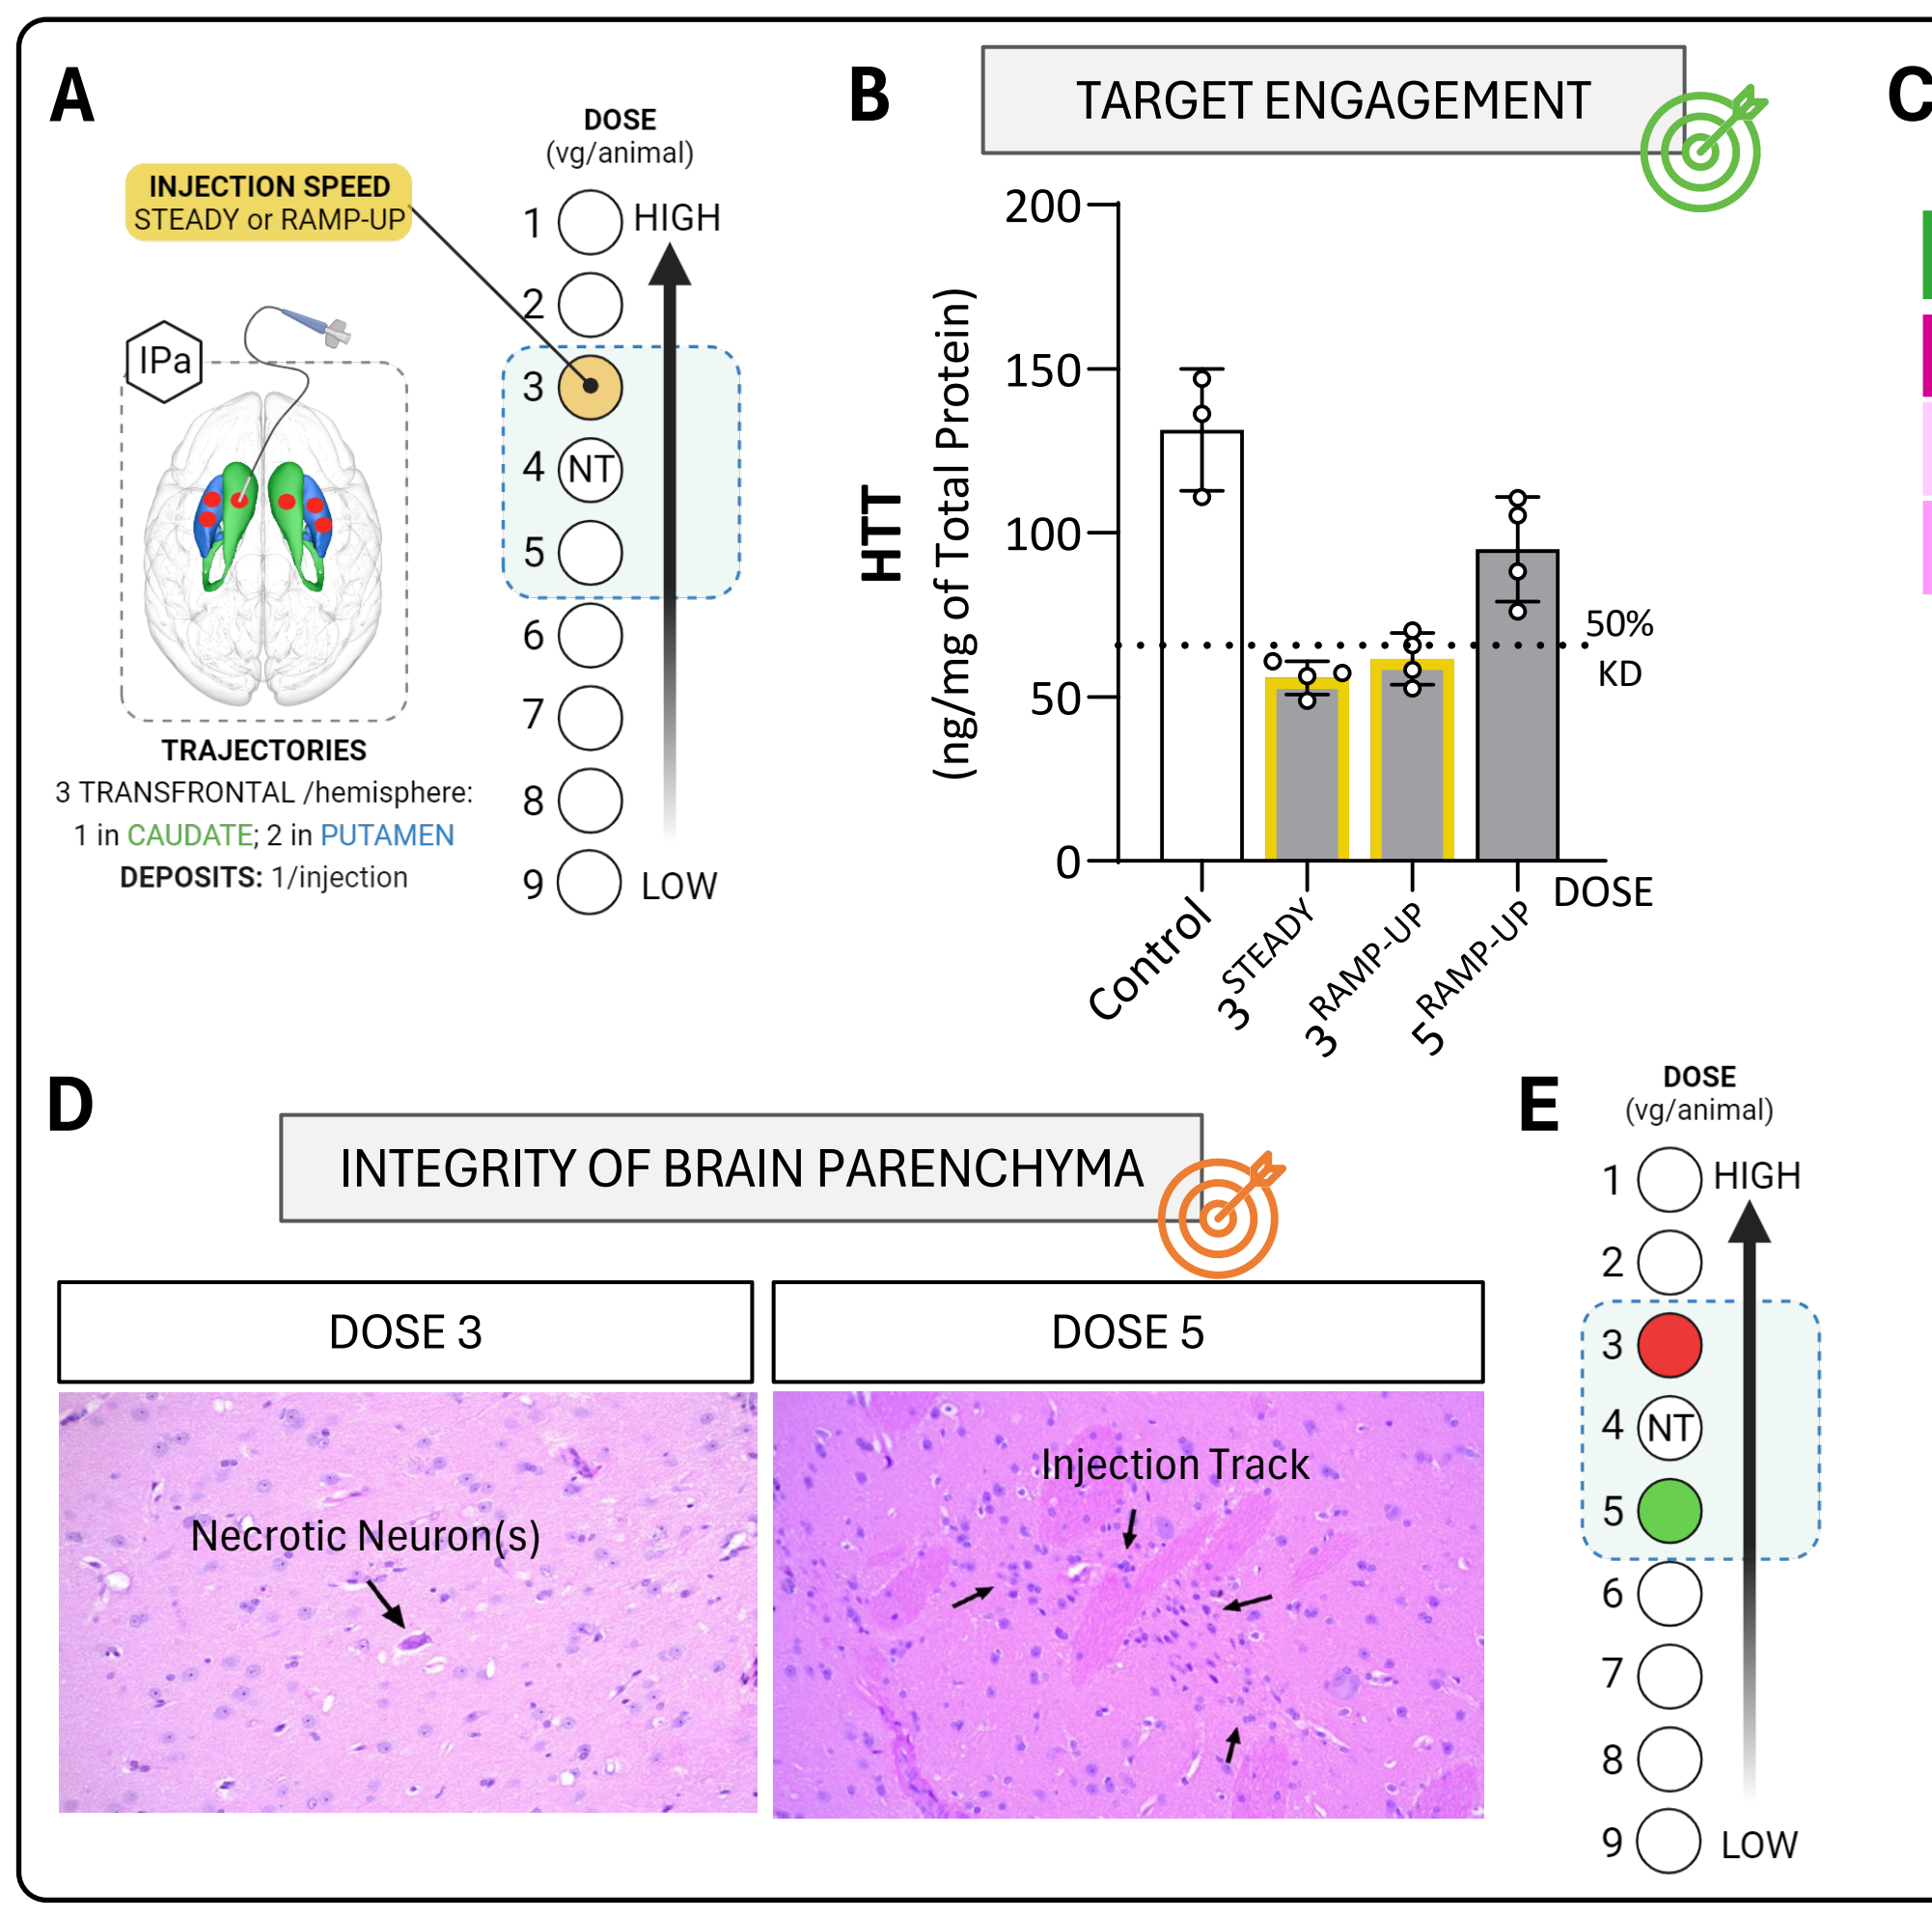
A
INJECTION SPEED
STEADY or RAMP-UP
DOSE (vg/animal)
HIGH
LOW
NT
TRAJECTORIES
3 TRANSFRONTAL /hemisphere:
1 in CAUDATE; 2 in PUTAMEN
DEPOSITS: 1/injection
B
TARGET ENGAGEMENT
HTT (ng/mg of Total Protein)
50% KD
Control
3 STEADY
3 RAMP-UP
5 RAMP-UP
DOSE
D
INTEGRITY OF BRAIN PARENCHYMA
DOSE 3
DOSE 5
Necrotic Neuron(s)
Injection Track
E
C
TARGET STRUCTURE COVERAGE

Average of Structure % Coverage (± SD)		
Injection Speed	caudate	putamen
RAMP-UP	19 ± 11	65 ± 7
STEADY	27 ± 16	68 ± 5

STUDY 3. INFUSION PARAMETERS SELECTION. IPa infusion of AAV formulation by ramp-up instead of steady speed increased safety while preserving AAV activity.
A. Schematic of the IPa injection procedure, the 2 doses (3 and 5, *blue box. NT=Not Tested*), and the 2 injection speeds (ramp-up or steady) tested in Study 3. n=4 animals/group. **B.** HTT protein in the putamen of cynomolgus macaques 3 months after injection. Dotted line indicates 50% protein knockdown (KD) relative to control (*white bar*). **C.** Post-injection MRI-scans analysis of Gadoteridol contrast media diffusion in the target structures. **D.** Representative images of the histopathology assessment of the putamen after IPa injection at two different speeds (10X objective magnification) **E.** Study 3 conclusions. *Red*= Effective dose but with adverse findings. *Green*= effective and safe dose.

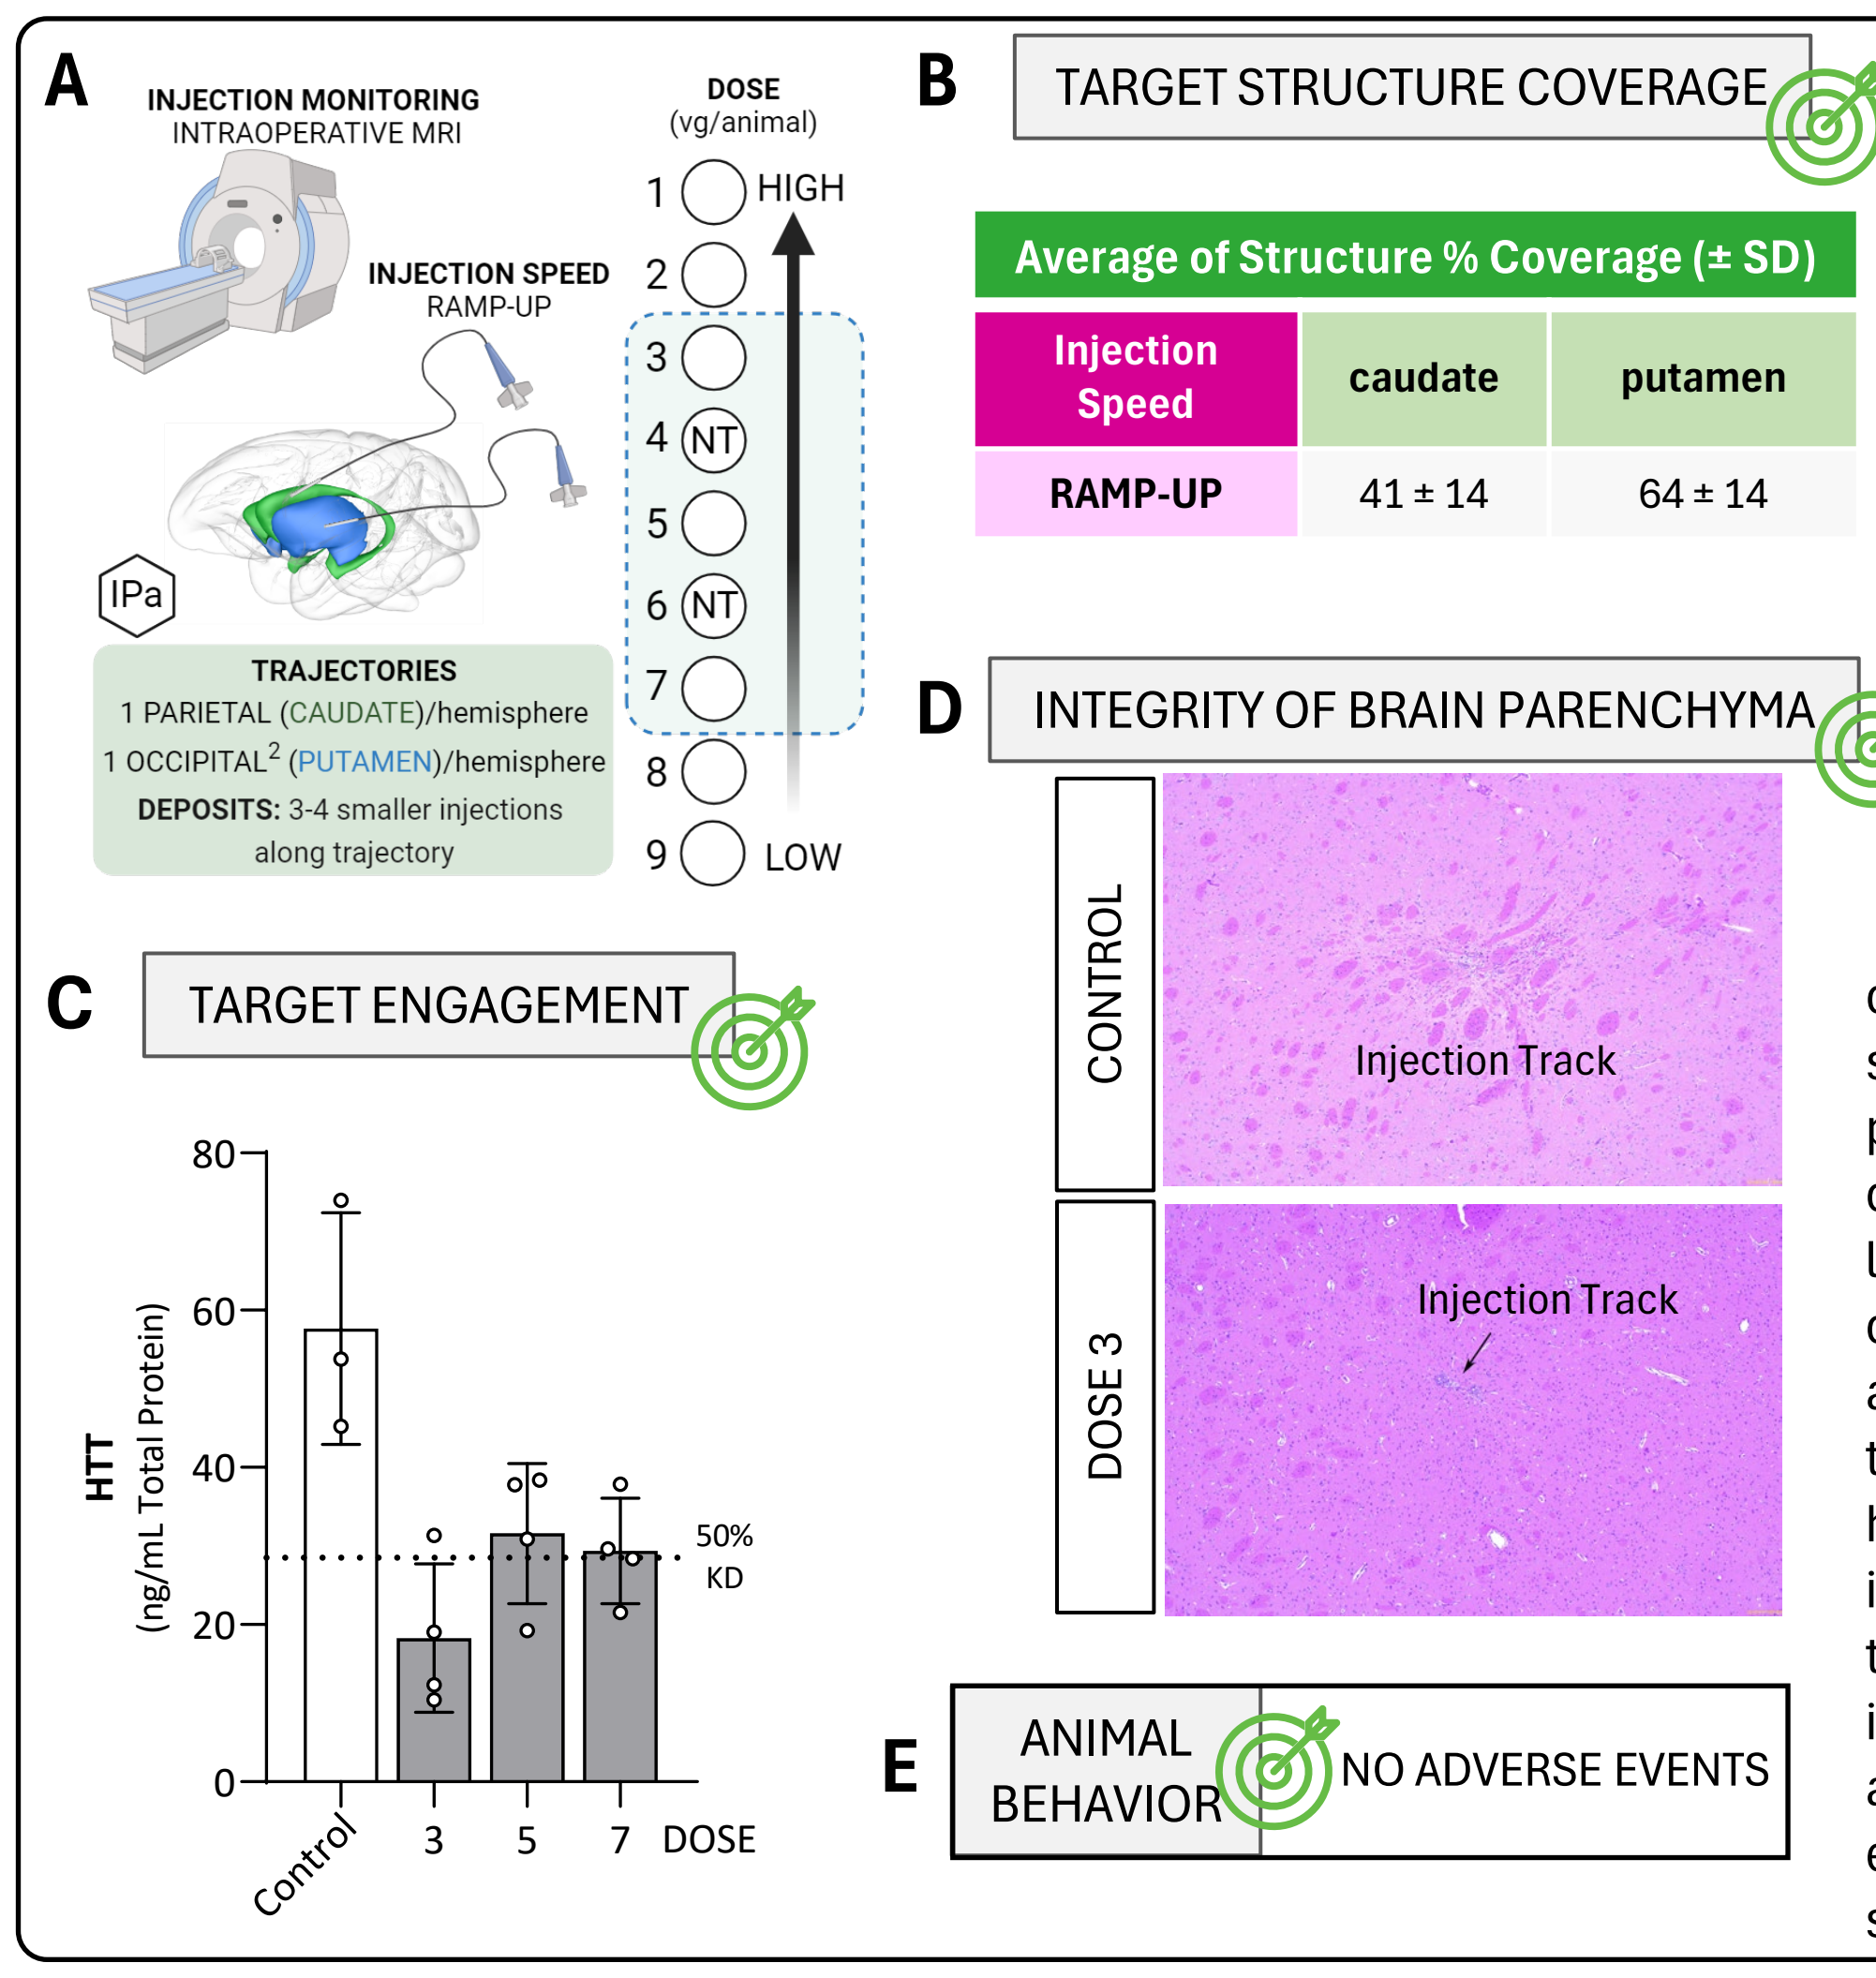
A
INJECTION MONITORING
INTRAOPERATIVE MRI
INJECTION SPEED
RAMP-UP
IPa
TRAJECTORIES
1 PARIETAL (CAUDATE)/hemisphere
1 OCCIPITAL2 (PUTAMEN)/hemisphere
DEPOSITS: 3-4 smaller injections along trajectory
DOSE (vg/animal)
HIGH
LOW
NT
C
TARGET ENGAGEMENT
HTT (ng/mL Total Protein)
50% KD
Control
DOSE
D
INTEGRITY OF BRAIN PARENCHYMA
CONTROL
DOSE 3
Injection Track
E
ANIMAL BEHAVIOR
NO ADVERSE EVENTS
F
B
TARGET STRUCTURE COVERAGE

Average of Structure % Coverage (± SD)		
Injection Speed	caudate	putamen
RAMP-UP	41 ± 14	64 ± 14

STUDY 4. TRAJECTORY SELECTION. Infusing the AAV formulation along parietal (caudate) and occipital (putamen) trajectories enhanced safety by minimizing the number of surgeries required per hemisphere while maintaining AAV activity.
A.Schematic of the IPa injection procedure under live-MRI monitoring, the 3 doses (3, 5, and 7, *blue box. NT=Not Tested*), and the 2 new trajectories tested in Study 4. n=4 animals/group. **B.** Post-injection MRI-scans analysis of Gadolinium contrast media diffusion in the target structures. caudate coverage is doubled compared to previous studies. **C.** HTT protein in the putamen of cynomolgus macaques 1.5 months after injection. Dotted line indicates 50% protein knockdown (KD) relative to control (*white bar*). Overall protein reduction is stable across studies despite the reduction in targeting trajectories. **D.** Representative images of the histopathology assessment of the putamen after IPa injection (4X objective magnification). Reduction of targeting trajectories couple with step-wise smaller infusions greatly improved safety of IP injections. **E.** No adverse events were associated with AAV delivery and expression. **F.** Study 4 conclusions. Green= effective and safe dose.

CONCLUSIONS

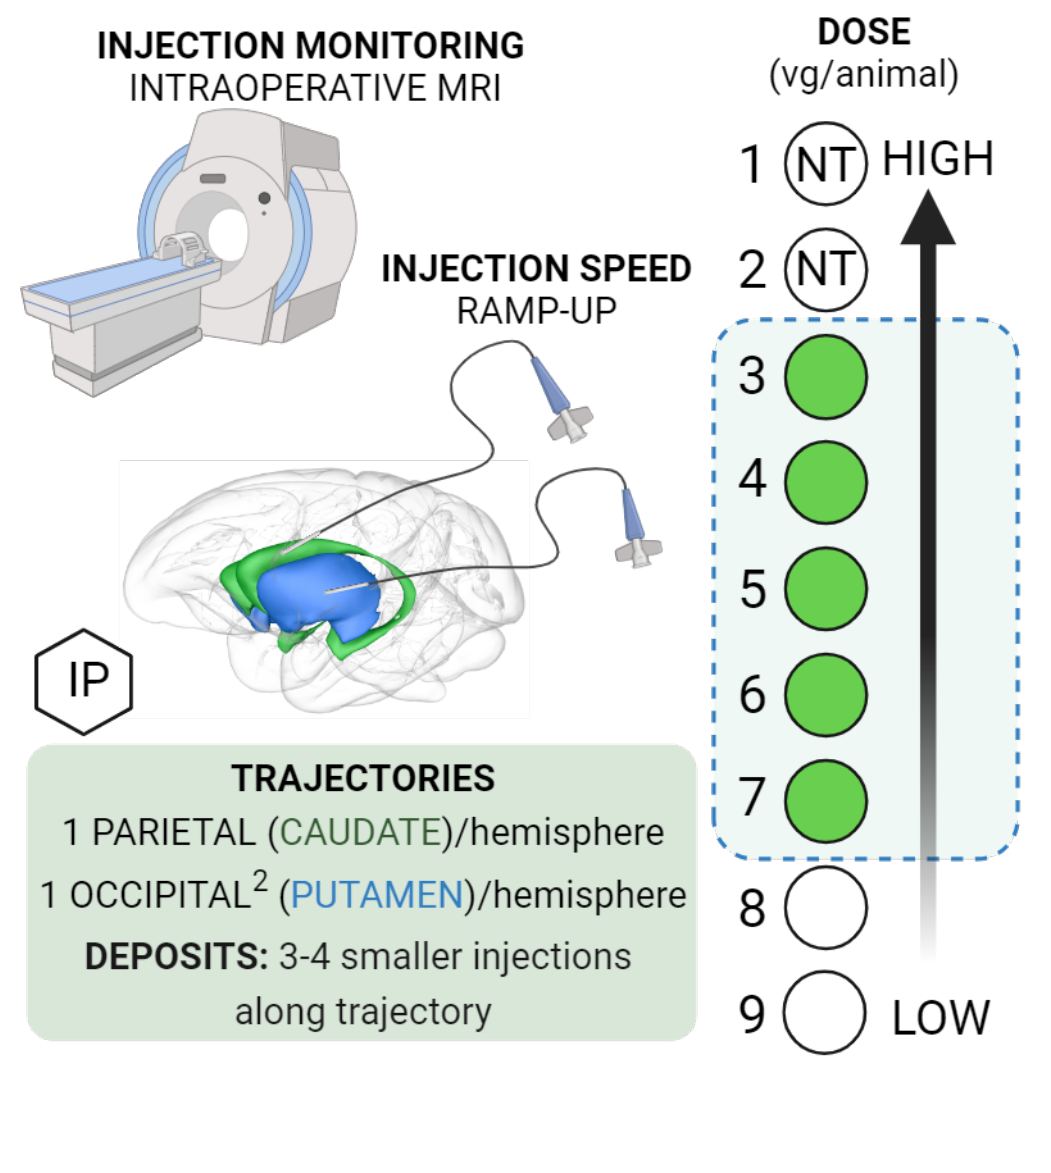
INJECTION MONITORING
INTRAOPERATIVE MRI
INJECTION SPEED
RAMP-UP
IP
TRAJECTORIES
1 PARIETAL (CAUDATE)/hemisphere
1 OCCIPITAL2 (PUTAMEN)/hemisphere
DEPOSITS: 3-4 smaller injections along trajectory
DOSE (vg/animal)
NT
HIGH
LOW

- **IPa delivery of AAV effectively reduces HTT protein in NHP caudate and putamen with minimal spread to the periphery and moderate increase of anti-AAV Nab titer compared to other routes (IT and ICV).**
- **Safety and efficacy of treatment depend on both AAV vector load and the surgical strategy that delivers them to the target brain structures**
- **The safest and most effective IPa strategy to deliver AAVs to caudate and putamen for Huntington's disease treatment is a step-wise release of the infusion in small deposits at gradually ramping speed along the longitudinal axis of the target structures under the monitoring of intraoperative MRI.**